F1-854
3038

Preclinical Toxicology of NP213, a Novel Fungicidal Peptide for the Treatment of Onychomycosis

323

Christopher A. Bamford, David B. Galloway & **Deborah A. O'Neil**

Background

Safety shortcomings of currently available therapeutics for fungal nail infections (onychomycosis) are a major driver for efforts to design, develop & introduce novel, improved treatment options to what remains a poorly served market. The efficacy potential of NP213, a novel topical peptide antifungal developed by NovaBiotics, has already been established *in vitro* and in *ex vivo* human nail models. A comprehensive preclinical safety & toxicology programme was then undertaken in order to confirm the potential of NP213 as a clinical candidate for a condition that affects as many as 12% of the global population.

Introduction

NP213 is a cyclic arginine heptamer (1093.3 Da) and the fungicidal API of Novexatin®, a clinical stage candidate therapeutic for the topical treatment of fungal nail infections (onychomycosis). Cationic antimicrobial peptides (cAMP) form the cornerstone of eukaryotic innate host defence, largely through broad-spectrum, membranolytic microbicidal action. The therapeutic application of these pleiotropic molecules is largely ruled out by a number of factors including physicochemcial characteristics & a pro-inflammatory & chemotactic capacity.

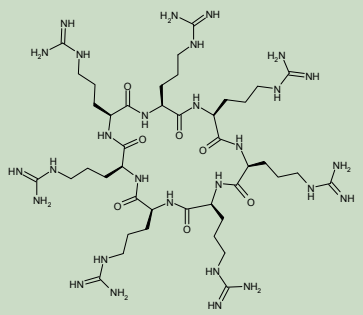

NP213 is a novel, third-generation synthetic cAMP with a size, charge, hydrophilicity & cyclic structure that facilitate penetration through the challenging biological barrier of the nail, but which prevents absorption through the skin. NP213 remains stable & bioavailable within the nail & is rapidly fungicidal against dermatophyte and other fungi causative of, or associated with onychomycosis therein (see poster F1-852).

NP213 provides a potential step-change solution to the as yet poorly met & economically significant clinical problem of onychomycosis. In line with regulatory requirements for a novel drug candidate intended for topical application in fungal nail infection, NP213 has been subjected to a comprehensive preclinical pharmacodynamic, toxicological & toxicokinetic programme to determine the molecule's suitability for clinical development.

Methods

In vitro tests were first performed to establish the cytotoxic & haemolytic potential of NP213. The data generated demonstrated no toxicity at doses significantly above antifungal MICs. Single & repeat dose rodent & non-rodent studies were next performed (by various partner CROs) in order to establish the safety & toxicological profile of NP213 *in vivo* by intravenous, oral & dermal routes. NP213 was tested in the proposed aqueous clinical formulation (PEG/Urea) in non-intravenous studies. Toxicokinetic tests were a component of the pivotal 28 day repeat exposure studies.

Pharmacology studies performed with NP213

STUDY	SYSTEM	KEY FINDINGS & OBSERVATIONS
Nail penetration	human/*ex vivo*	Penetration into & through full thickness intact nail assessed by mycology, ELISA and radiochemical assay
Skin penetration	human/*ex vivo*	Dermal delivery of only 0.03% NP213, absorbed dose 0.01%
Eye irritation	rodent/*in vivo*	Minor & transient, self resolving irritation only
Dermal sensitisation	rodent/*in vivo*	No sensitisation
	mini-pig/*in vivo*	No sensitisation

Single-dose toxicity studies performed with NP213

Rat, intravenous, doses up to 20 mg/kg, 15 days observation	NOAEL >1 mg/kg, MTD 1-3 mg/kg
Mouse, oral, 200-500 mg/kg dosed, 3 days observation	MTD 250-280 mg/kg
Mini-pig dermal MTD/dose-range study. 14 days observation	MTD >100 mg/kg

Toxicokinetics

28 day rat intravenous study	- no accumulation of test compound seen with repeat dosing - CL lower than known renal and hepatic flow rates - high Vd: extensive distribution beyond central circulation - t1/2 = 3-6hrs - no sex related differences
Dosing 1.25 mg/kg/day, Cmax at day 28 of 6523 ng/ml & AUC (h*ng/ml) of 31138	

Repeat exposure studies performed with NP213

STUDY	KEY FINDINGS & OBSERVATIONS
7 day rat intravenous MTD/acute toxicity study, dosing up to 4 mg/kg/day	NOAEL = 0.5 mg/kg/day in males, 1.25 mg/kg/day in females MTD = 2 mg/kg/day,
14 day canine intravenous MTD study, dosing up to 4 mg/kg/day	MTD = 2 mg/kg/day
Pivotal Rat 28 day intravenous study	As described in toxicokinetic study summary
Pivotal 28 day mini-pig toxicokinetic study dermal daily application of up to 50 mg/kg NP213	No observed tolerability issues or sensitisation No systemic exposure even at highest doses applied NOAEL >50 mg/kg/day

Results

Safety margins established for NP213 in relation to the proposed clinical dose (Novexatin® contains 10% (w/v) NP213 in PEG/Urea) were 227 x from a pivotal 28 day repeat dermal exposure mini-pig study & >400 x from a rat intravenous 28 day repeat exposure study. No sensitisation was observed in mini-pig & rodent dermal challenge studies & favourable rodent oral toxicology & eye irritation profiles were obtained. There was no test item related toxicity in any mini-pig studies & toxicokinetic analyses demonstrated no systemic exposure (now also confirmed in patients in phase 1 of clinical development). The plasma half-life for NP213 was determined as 2-3 hours from a pivotal rat intravenous study.

Conclusions

NP213 has an encouraging safety & toxicological profile in line with clinical development plans for the therapeutic application of an antifungal peptide as the API of Novexatin®, a novel, rapidly acting, 28 day topical treatment for onychomycosis. Phase 1 of clinical development for Novexatin® is now complete & a 28 day repeat dose phase 2a component of this first time in man study is scheduled to conclude in 2009.

info@novabiotics.co.uk
www.novabiotics.co.uk

NovaBiotics Ltd®
Cruickshank Building, Craibstone
Aberdeen, AB21 9TR, UK

☎ 0044 (0)1224 711377
0044 (0)1224 711370